A model-based meta-marker to characterize the response to ranibizumab in wet AMD patients

Cheikh Diack, Dietmar Schwab, Norman Mazer, Vincent Buchheit, Valerie Cosson, Nicolas Frey

Roche Pharma Research and Early Development, Clinical Pharmacology, Roche Innovation Center Basel
F. Hoffmann-La Roche Ltd, Switzerland

1 – Introduction and objectives

Ranibizumab (Lucentis®) is an anti-vascular endothelial growth factor (VEGF) antibody fragment used in the treatment of wet age-related macular degeneration (AMD). The disease is characterized by a rapid decline in visual acuity (VA) over the course of months while treatment with ranibizumab results on average in an improvement of VA. A drug-disease model was developed to describe the changes over time in VA of patients under Lucentis injections and the progression of the disease in terms of VA loss for patients receiving a sham treatment.

Objectives: To derive a composite parameter (meta-marker) from the baseline characteristics that quantitates the high heterogeneity in response to ranibizumab and in deterioration of vision in wet AMD.

2 – Methods

Data

1) Visual acuity data from the ANCHOR, MARINA and HARBOR trials of ranibizumab were available (n=2243 patients with baseline characteristics, including 240 sham patients).
2) All available data (Fig 1 and Fig 2) were used to developed the structural model.
3) Baselines covariates from fundus fluorescence angiography (FFA) and from optical coherence tomography (OCT) were available

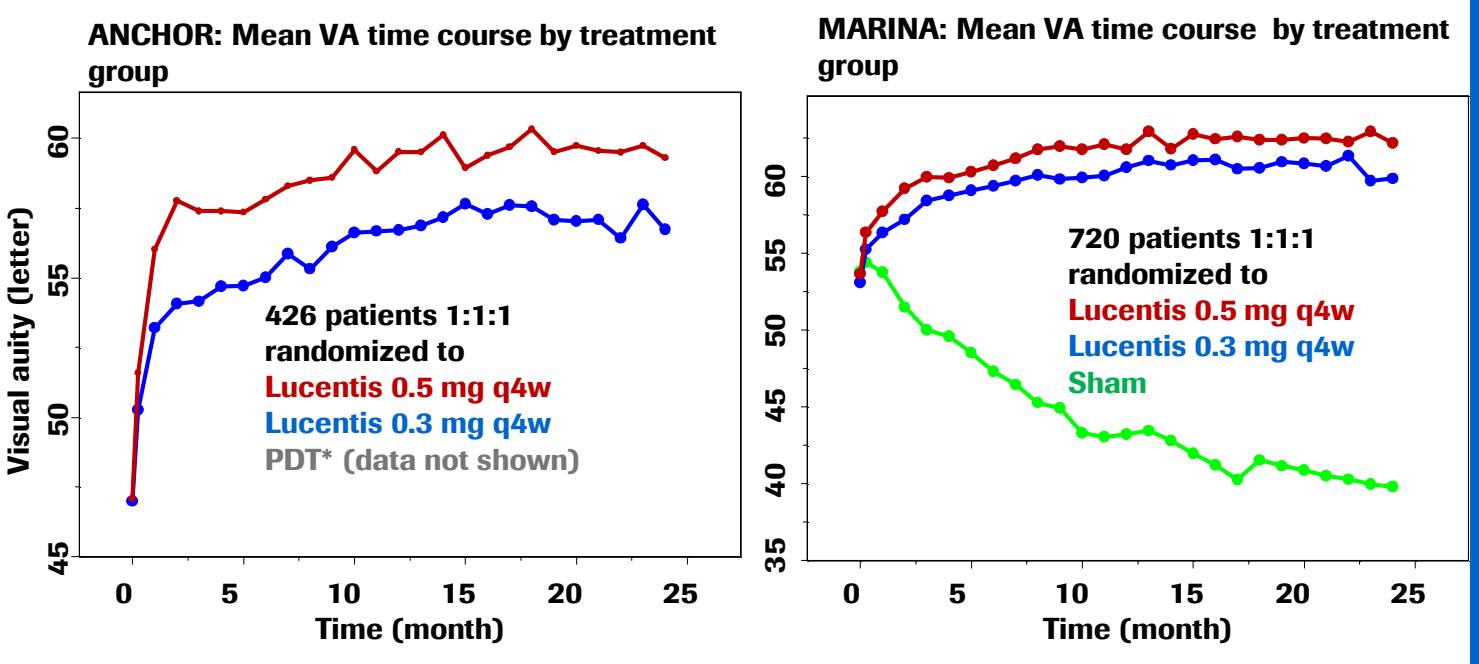
ANCHOR: Mean VA time course by treatment group
Visual auity (letter)
45
50
55
60
0
5
10
15
20
25
Time (month)
426 patients 1:1:1
randomized to
Lucentis 0.5 mg q4w
Lucentis 0.3 mg q4w
PDT* (data not shown)
MARINA: Mean VA time course by treatment group
35
40
45
50
55
60
0
5
10
15
20
25
Time (month)
720 patients 1:1:1
randomized to
Lucentis 0.5 mg q4w
Lucentis 0.3 mg q4w
Sham

Fig 1: VA mean profiles of data used for the structural model

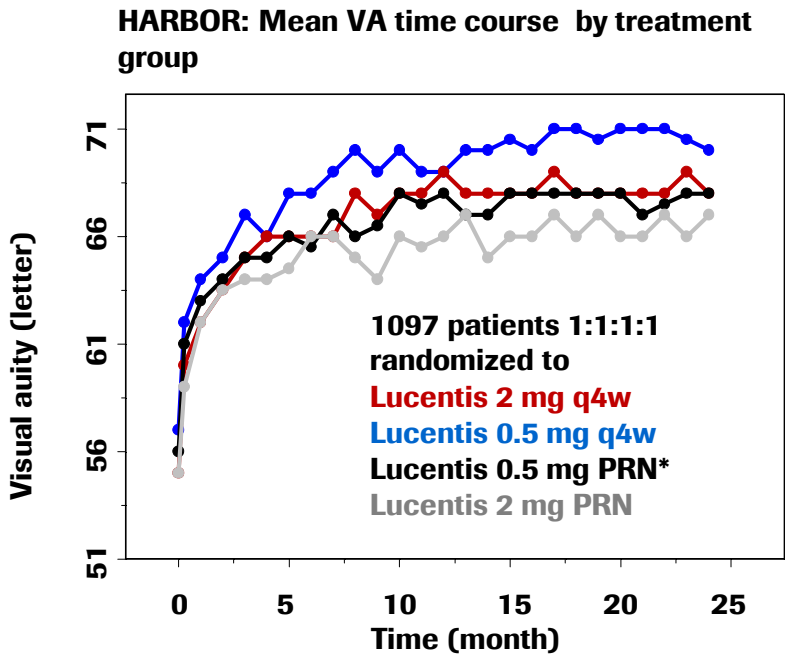
HARBOR: Mean VA time course by treatment group
Visual auity (letter)
51
56
61
66
71
0
5
10
15
20
25
Time (month)
1097 patients 1:1:1:1
randomized to
Lucentis 2 mg q4w
Lucentis 0.5 mg q4w
Lucentis 0.5 mg PRN*
Lucentis 2 mg PRN

Fig 2: VA mean profiles of data used for the structural and covariate models

Models

Adaptations of the previous structural model in [1] were made : 1) uncertainty related to baseline VA was estimated but not baseline VA; 2) baseline VA was a covariate of the maximum effect and of VA at steady when the patient was not under treatment (cf. equations 1 and 2).

(1) $$VA(t) = VA_0 - (VA_0 - VA_{ss}) \cdot \left(1 - e^{-kpr\left(1-\frac{IR}{Ed50+IR}\right).t}\right) + \frac{E\max.IR}{E50+IR}$$

(2) $$VA_{ss} = VA_{ss0} * \left(\frac{VA_0}{100}\right)^p$$ **and** $$E\max = E\max{}_0 * \left(1 - \left(\frac{VA_0}{100}\right)^q\right)$$

VA (t): VA at time *t*; **VA0:** VA at baseline; **VAss:** VA at steady-state when the patient is not under treatment; ***IR:*** the input rate of drug at time *t*; ***kpr*** the rate of disease progression.

The model assumes two effects of Lucentis: a modification of disease progression (effect on *kpr*) and a symptomatic effect (additive Emax) .
The influence of available baseline characteristics on model parameters were tested

Population data analysis

The data were modeled using a nonlinear mixed effects modeling approach in NONMEM (version 7.2 Icon Development Solutions, Ellicott City, MD) for the parameters estimation and SPLUS (version 8.0; Insightful, Seattle, WA) was used for graphical analyses of the results.
Residual variability was modeled with an additive error model.
Model development and qualification were guided by the objective function (OFV), the goodness of fit plots (GOF) and the precision of parameter estimates. Several baseline covariates from fundus fluorescence angiography (FFA) and from optical coherence tomography (OCT) were tested on model parameters.

*** PRN: Injections monthly for 3 months then as needed.**
*** PDT: photodynamic therapy**

3 – Results

Model-based meta-marker (MBMM)

The final model is defined by equations (1) and (2) with baseline covariates on model parameters *Kpr Emax* and E50. The identified covariates (which cannot be reported at this time) were found to be plausible from a pathophysiologic context. Goodness-of-fit plots and visual predictive checks (not shown) suggests that the model can describe adequately the data.

For each wet AMD patient a MBMM was defined as the population projected change from baseline of VA at 2 years when treated with Lucentis.

Figure 3 shows the distribution of the MBMM for the 0.5 mg q4w arm of the HARBOR study.
The distrubtion in Figure 3 is used as a reference distribution to characterize responses to ranibizumad in wet AMD patients: **poor responders** are defined as the patients with MBMM lower than the 30th percentile while **super responders** are patients with MBMM igreater than or equal the 70th percentile.

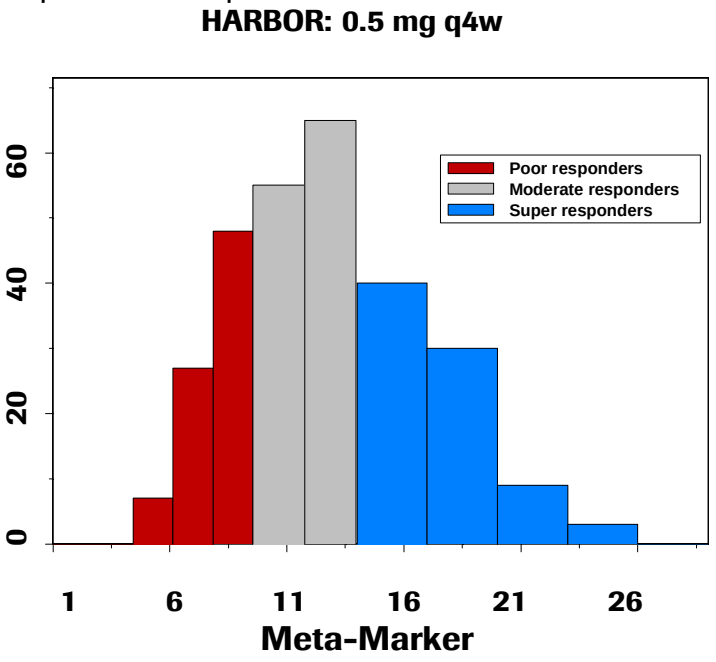
HARBOR: 0.5 mg q4w
0
20
40
60
Poor responders
Moderate responders
Super responders
1
6
11
16
21
26
Meta-Marker

Fig 3: MBMM distribution

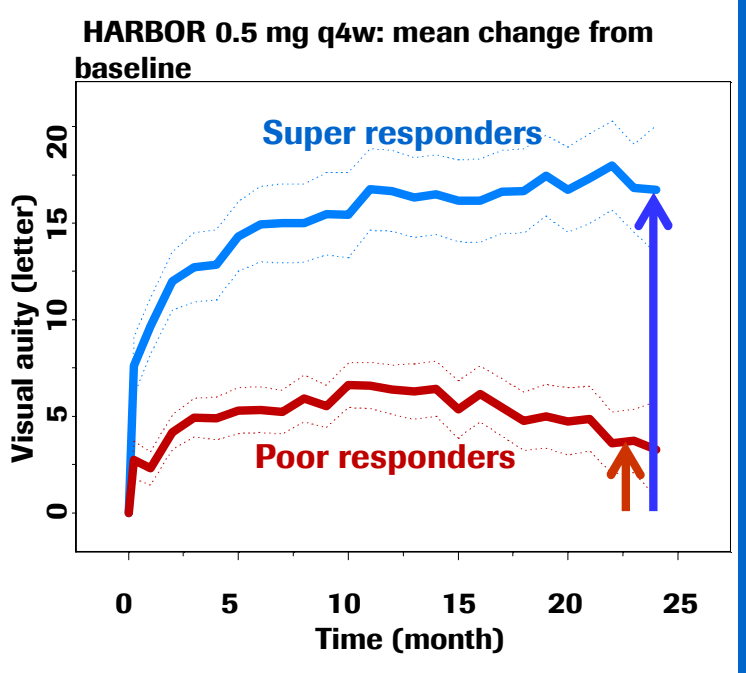
HARBOR 0.5 mg q4w: mean change from baseline
Visual auity (letter)
0
5
10
15
20
Super responders
Poor responders
0
5
10
15
20
25
Time (month)

Fig 4: VA observed mean change over time

Figure 4 shows the observed means change from baseline of VA for each of these groups of responders **(16 letters at month 12 for super responders** **and** **6 letters for poor responders)**
The methods used here were successfully applied to the other dose groups from HARBOR (data not shown)

Assessing responders from the progress state

As above, responders are usually assessed by their change from baseline. This does not take into account the likely difference in disease progression between patients.
<u>New hypothesis:</u> «apparent» super responders and poor responders are more similar drug effects when assessed from their progressed state as shown in Figure 5.

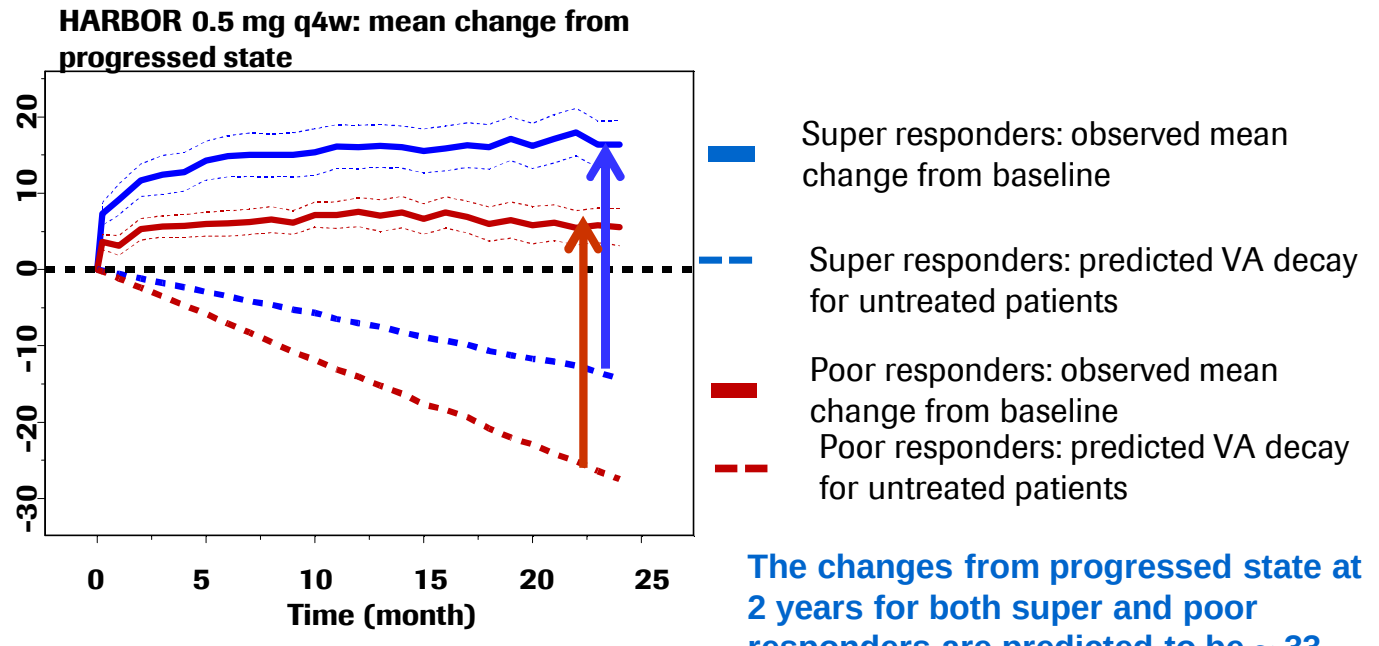
HARBOR 0.5 mg q4w: mean change from progressed state
-30
-20
-10
0
10
20
0
5
10
15
20
25
Time (month)
Super responders: observed mean change from baseline
Super responders: predicted VA decay for untreated patients
Poor responders: observed mean change from baseline
Poor responders: predicted VA decay for untreated patients

Fig 5

The changes from progressed state at 2 years for both super and poor responders are predicted to be ~ 33 letters.

4 – Conclusions

The proposed model-based meta-marker proves to be an important factor in explaining the heterogeneity in response to ranibizumab treatment.
In all examples tested so far, the MBMM has provided good prediction at the population level.

Based on the model we also suggest that the main difference between "poor" and "super' responders is that the former would have a much greater deterioration in VA if left untreated than the latter patient population.

<u>References:</u>

[1] Diack C., Schwab D., Frey N. **An empirical drug-disease model to characterize the effect of Ranibizumab on disease progression in wet AMD patients.** ***PAGE 24 (2015) Abstr 3569 [www.page-meeting.org/?abstract=3569]***